Menin Inhibition in Pediatric *KMT2Ar* Leukemia: Phase 1 and Expanded Access Experience With Revumenib

108

Branko Cuglievan,[1] David Dickens,[2] Shalini Shenoy,[3] Jeffrey Rubnitz,[4] Raul Ribeiro,[4] Andrew E. Place,[5] Naval Daver,[1] Irina Zaidman,[6] Pinki Prasad,[7] Anna M. Ewins,[8] Nicole McNeer,[9] Yu Gu,[9] Soujanya Sunkaraneni,[9] Rachel Ghiraldi,[9] Neerav Shukla[10]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]University of Iowa Stead Family Children's Hospital, Iowa City, IA, USA; [3]Washington University School of Medicine Siteman Cancer Center, St. Louis, MO, USA; [4]St. Jude Children's Research Hospital, Memphis, TN, USA; [5]Dana-Farber/Boston Children's Cancer and Blood Disorder Center, Boston, MA, USA; [6]Hadassah-Hebrew University Medical Center, Jerusalem, Israel; [7]Louisiana State University Health and Children's Hospital of New Orleans, New Orleans, LA, USA; [8]The Royal Hospital for Children, Glasgow, Scotland, UK; [9]Syndax Pharmaceuticals, Inc, Waltham, MA, USA; [10]Memorial Sloan Kettering Cancer Center, New York, NY, USA

Presenting Author: Branko Cuglievan; Bcuglievan@mdanderson.org

INTRODUCTION

- Acute leukemias with histone-lysine N-methyltransferase 2A rearrangements (*KMT2Ar*, previously *MLLr*) are hematopoietic malignancies with a poor prognosis that account for the majority of acute leukemias diagnosed before the age of 2 years[1,2]
 - *KMT2Ar* accounts for 15% to 20% of pediatric acute myeloid leukemia cases and 5% to 6% of pediatric acute lymphoblastic leukemia cases[3,4]
- Disrupting the KMT2A-menin interaction in preclinical models leads to downregulation of *HOX/MEIS1* genes and reversal of leukemogenesis[5,6]
- Revumenib (SNDX-5613) is an investigational, potent, selective inhibitor of the KMT2A-menin interaction, with demonstrated preliminary antileukemic activity in the phase 1 AUGMENT-101 (NCT04065399) study in patients with relapsed/refractory (R/R) acute *KMT2Ar* and *NPM1c* leukemias[6]

OBJECTIVE

- To evaluate revumenib treatment in pediatric patients with R/R acute *KMT2Ar* and *NPM1c* leukemias in the AUGMENT-101 study and to provide access to patients not eligible for a clinical trial via single patient protocols

METHODS

AUGMENT-101 Study Patients (Data cutoff March 31, 2022)

- The AUGMENT-101 phase 1 dose escalation has been previously reported[6]
- 8 patients aged <18 years were enrolled and received either revumenib without (arm A) or with (arm B) concomitant strong cytochrome P450 3A4 (CYP3A4) inhibitors
 - All 8 pediatric patients received a dose that met the criteria for the recommended phase 2 dose
- Adverse events (AEs) were collected and reported per protocol and central review of efficacy was conducted

Revumenib Expanded Access Patients (Data cutoff December 22, 2022)

- 20 patients ineligible for AUGMENT-101 who had exhausted all other approved treatment options were granted expanded access to revumenib with or without concomitant strong CYP3A4 inhibitors via single patient protocols
 - Reasons for ineligibility included: young age (before protocol amendment) or disease status (eg, uncontrolled infection, poor performance status, presence of central nervous system disease)
 - 1 patient received revumenib as maintenance therapy after previous exposure
- In patients <40 kg, body surface area (BSA)-based dosing was implemented according to standard allometric scaling and subsequently supported by pharmacokinetic data from AUGMENT-101
- Efficacy and AEs were reported at the physician's discretion

1. AUGMENT-101 trial schema

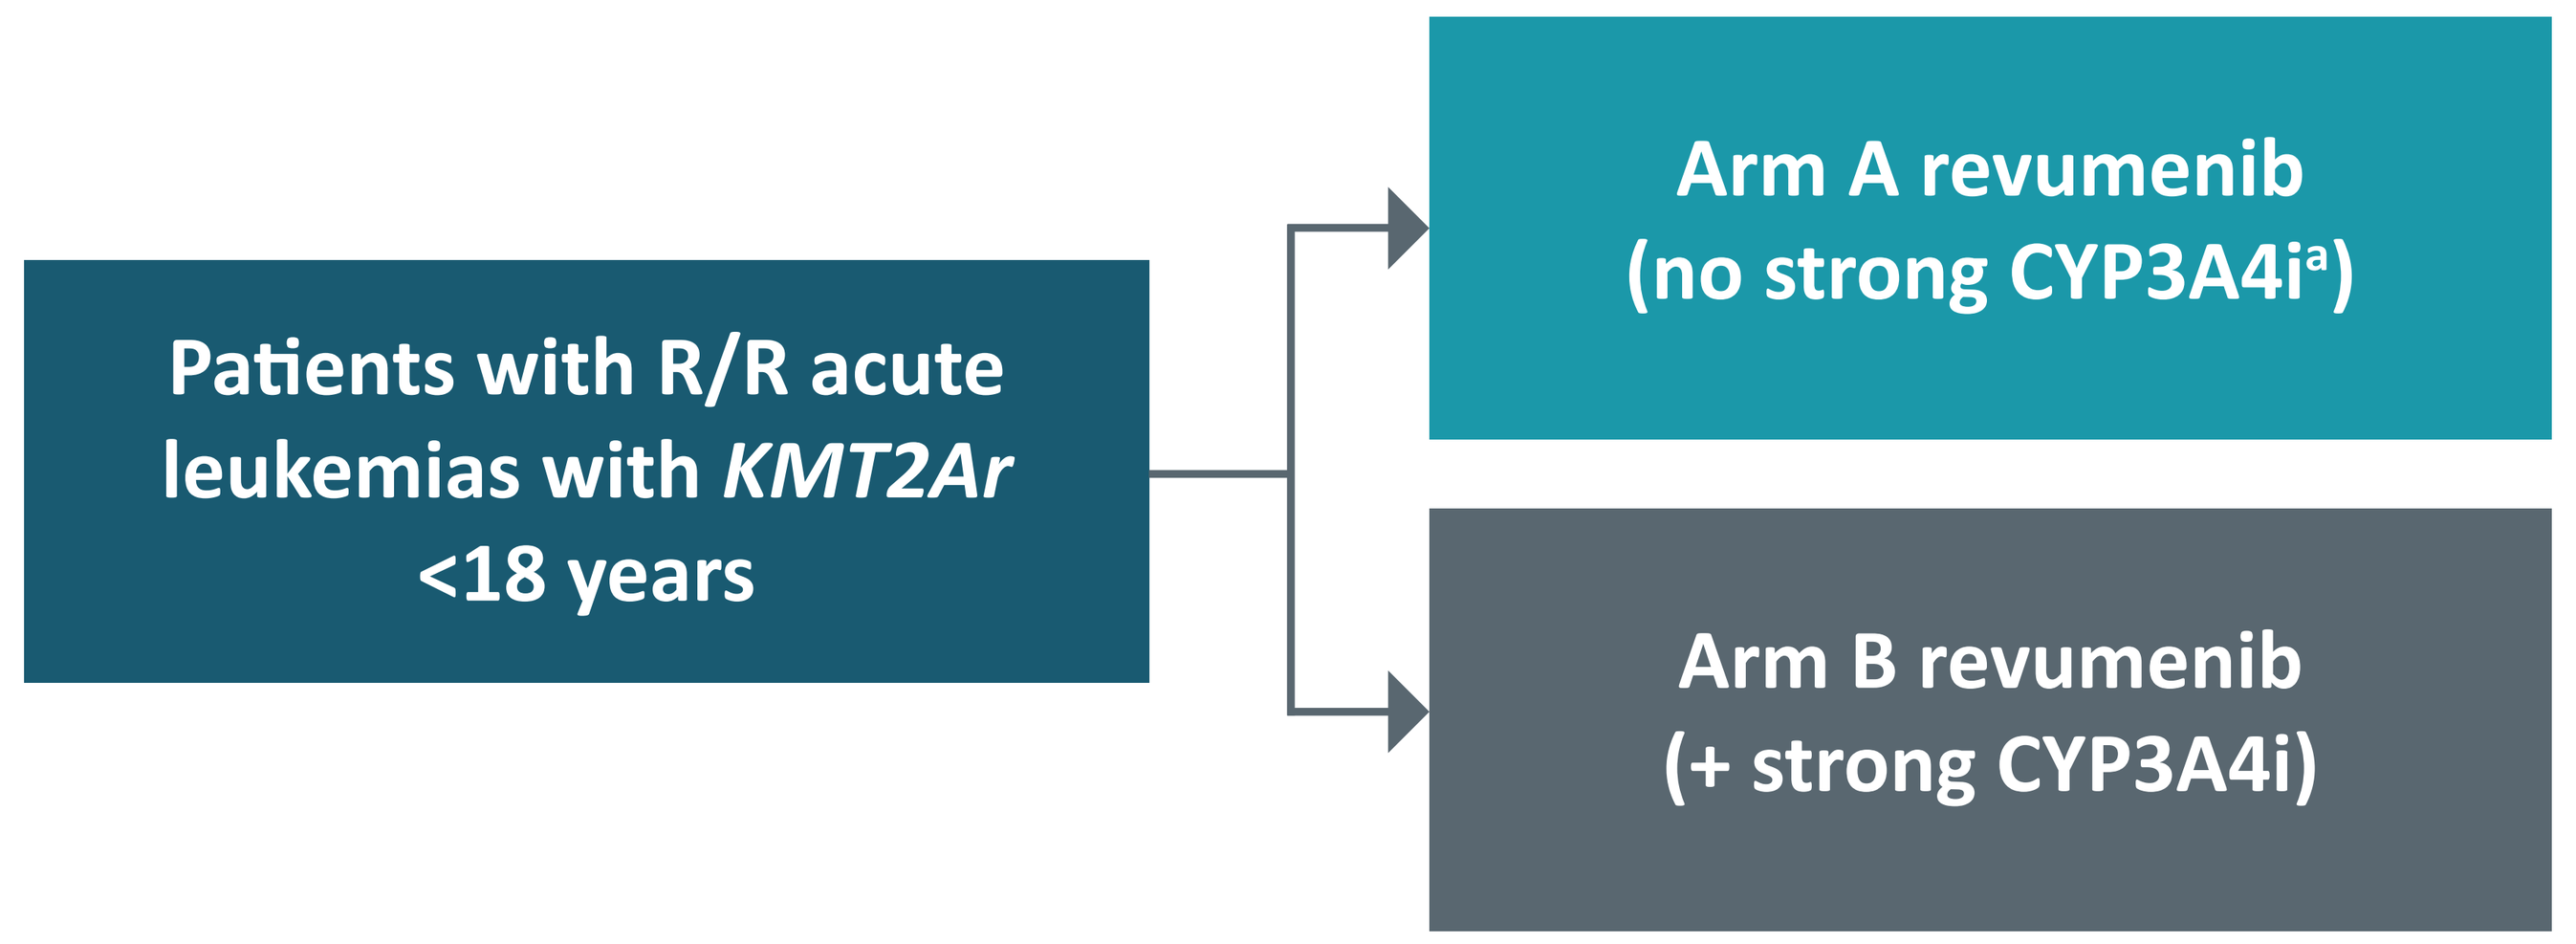
Patients with R/R acute leukemias with *KMT2Ar* <18 years
Arm A revumenib (no strong CYP3A4i[a])
Arm B revumenib (+ strong CYP3A4i)

CYP3A4i, cytochrome P450 3A4 inhibitor; R/R, relapsed/refractory. [a]No strong CYP3A4i = none + moderate CYP3A4i

RESULTS

BASELINE CHARACTERISTICS

- Patients with R/R *KMT2Ar* acute leukemia (n=8) who enrolled in arms A or B of AUGMENT-101 were aged 9 months to 16 years
 - 25% of patients had ≥5 prior lines of therapy and 50% had prior hematopoietic stem cell transplant (HSCT)
- Patients with R/R *KMT2Ar* leukemia (n=20) who received revumenib through an expanded access program were aged 14 months to 17.9 years
 - 30% of patients had ≥5 prior lines of therapy and 55% of patients had prior HSCT

Patient Demographics and Baseline Characteristics

Parameter	AUGMENT-101 (n=8)	Expanded access group (n=20)
Median age, y (range)	2.5 (0.8-16.0)	7.0 (1.2-17.9)
Female, n (%)	5 (62.5)	10 (50.0)
Male, n (%)	3 (37.5)	10 (50.0)
Strong CYP3A4i therapy, n (%)	1 (12.5)	11 (55.0)
Number of prior lines, median of therapy (range)	5 (2-10)	4 (1-6)
≥5 prior lines of therapy, n (%)	2 (25.0)	6 (30.0)
Prior HSCT, n (%)	4 (50.0)	11 (55.0)

CYP3A4i, cytochrome P450 3A4 inhibitor; HSCT, hematopoietic stem cell transplant.

EFFICACY

AUGMENT-101

- The ORR for patients treated in AUGMENT-101 was 50% (4/8)
 - 1 patient experienced complete response with partial hematologic recovery (CRh) and negative minimal residual disease (MRD) status, 1 patient achieved complete response with incomplete platelet recovery (CRp), and 2 patients experienced morphologic leukemia-free state
 - The patients with CRh and CRp received subsequent HSCT at data cutoff
 - Progressive disease (PD) and no response were observed in 2 patients each

2. Best overall response for patients from AUGMENT-101

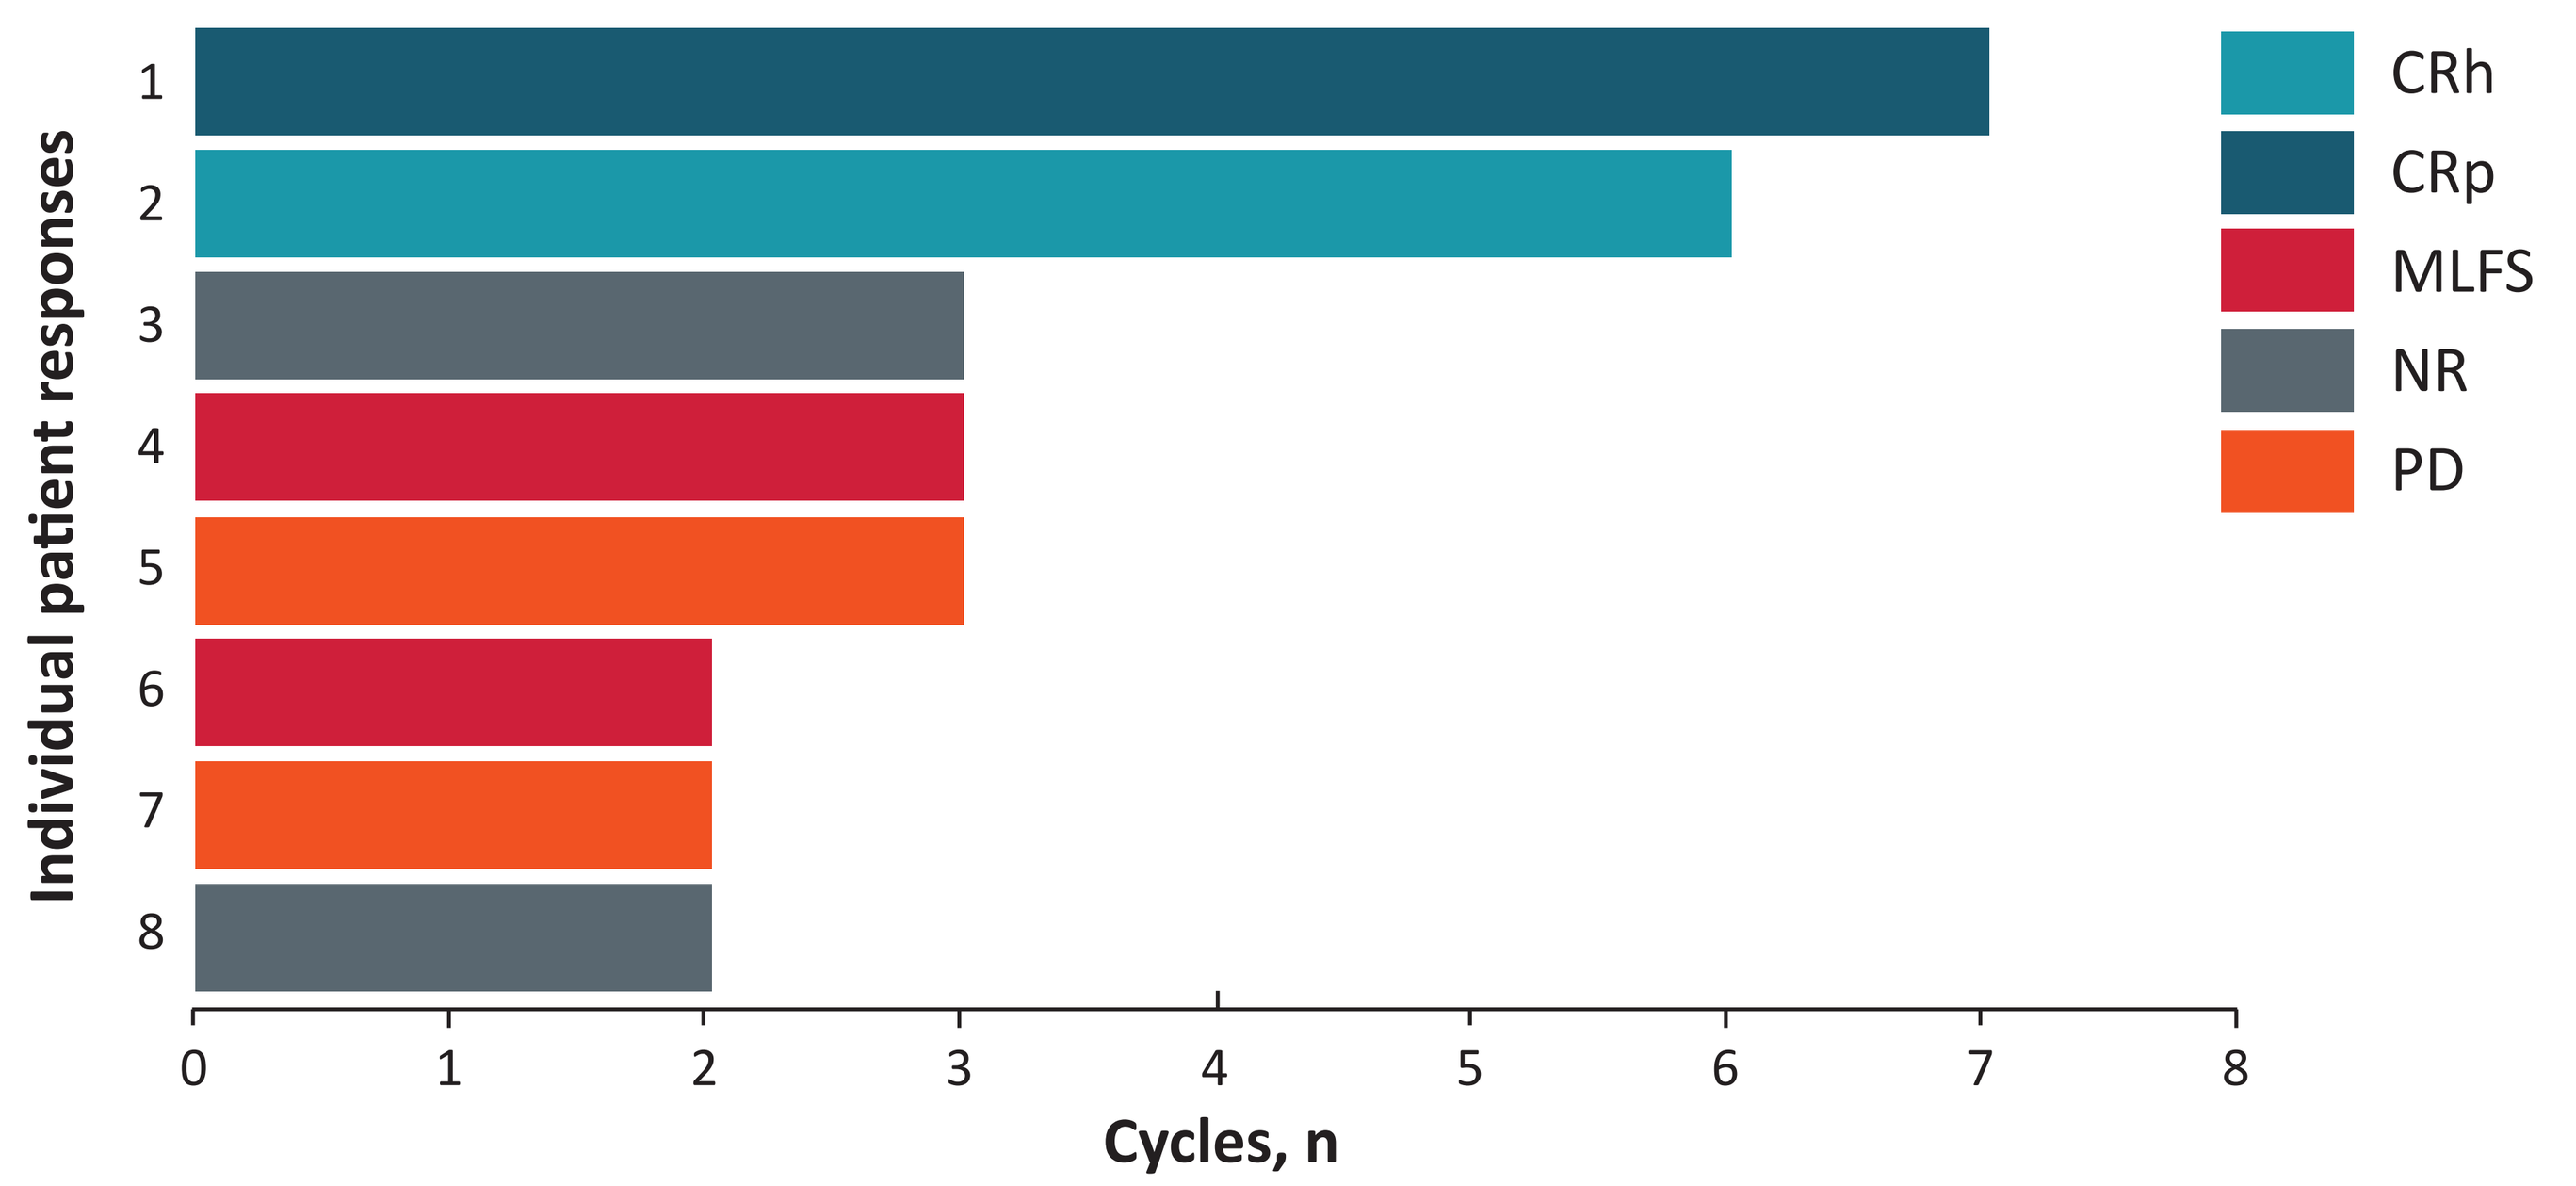
Individual patient responses
1
2
3
4
5
6
7
8
0 1 2 3 4 5 6 7 8
Cycles, n
CRh
CRp
MLFS
NR
PD

Swimmer plot illustrating the best overall response of the 8 pediatric patients treated with revumenib in the AUGMENT-101 trial. CRh, complete response with partial hematologic recovery; CRp, complete response with incomplete platelet recovery; MLFS, morphologic leukemia-free state; NR, no response; PD, progressive disease.

Revumenib Expanded Access

- A total of 4 patients experienced complete response (CR) with revumenib treatment in the expanded access group, of which:
 - 3 patients achieved CR (2 achieved MRD negative status) and 1 achieved CR with incomplete hematologic recovery (CRi)
 - Both patients who achieved CR with negative MRD status and the patient with CRi proceeded to receive HSCT
- 1 patient who achieved CR in AUGMENT-101 received revumenib as post-HSCT maintenance therapy
 - Patient has remained on treatment and in CR as of day 158 of therapy and was reported clinically well

PHARMACOKINETICS

- For patients in the AUGMENT-101 trial, dose and exposure followed a typical allometric relationship, with no notable differences compared with adults
- BSA-based dosing strategy for pediatric patients yields exposure similar to that of adults

3. BSA-based dosing strategy of revumenib in pediatric patients yields exposure similar to their adult counterparts

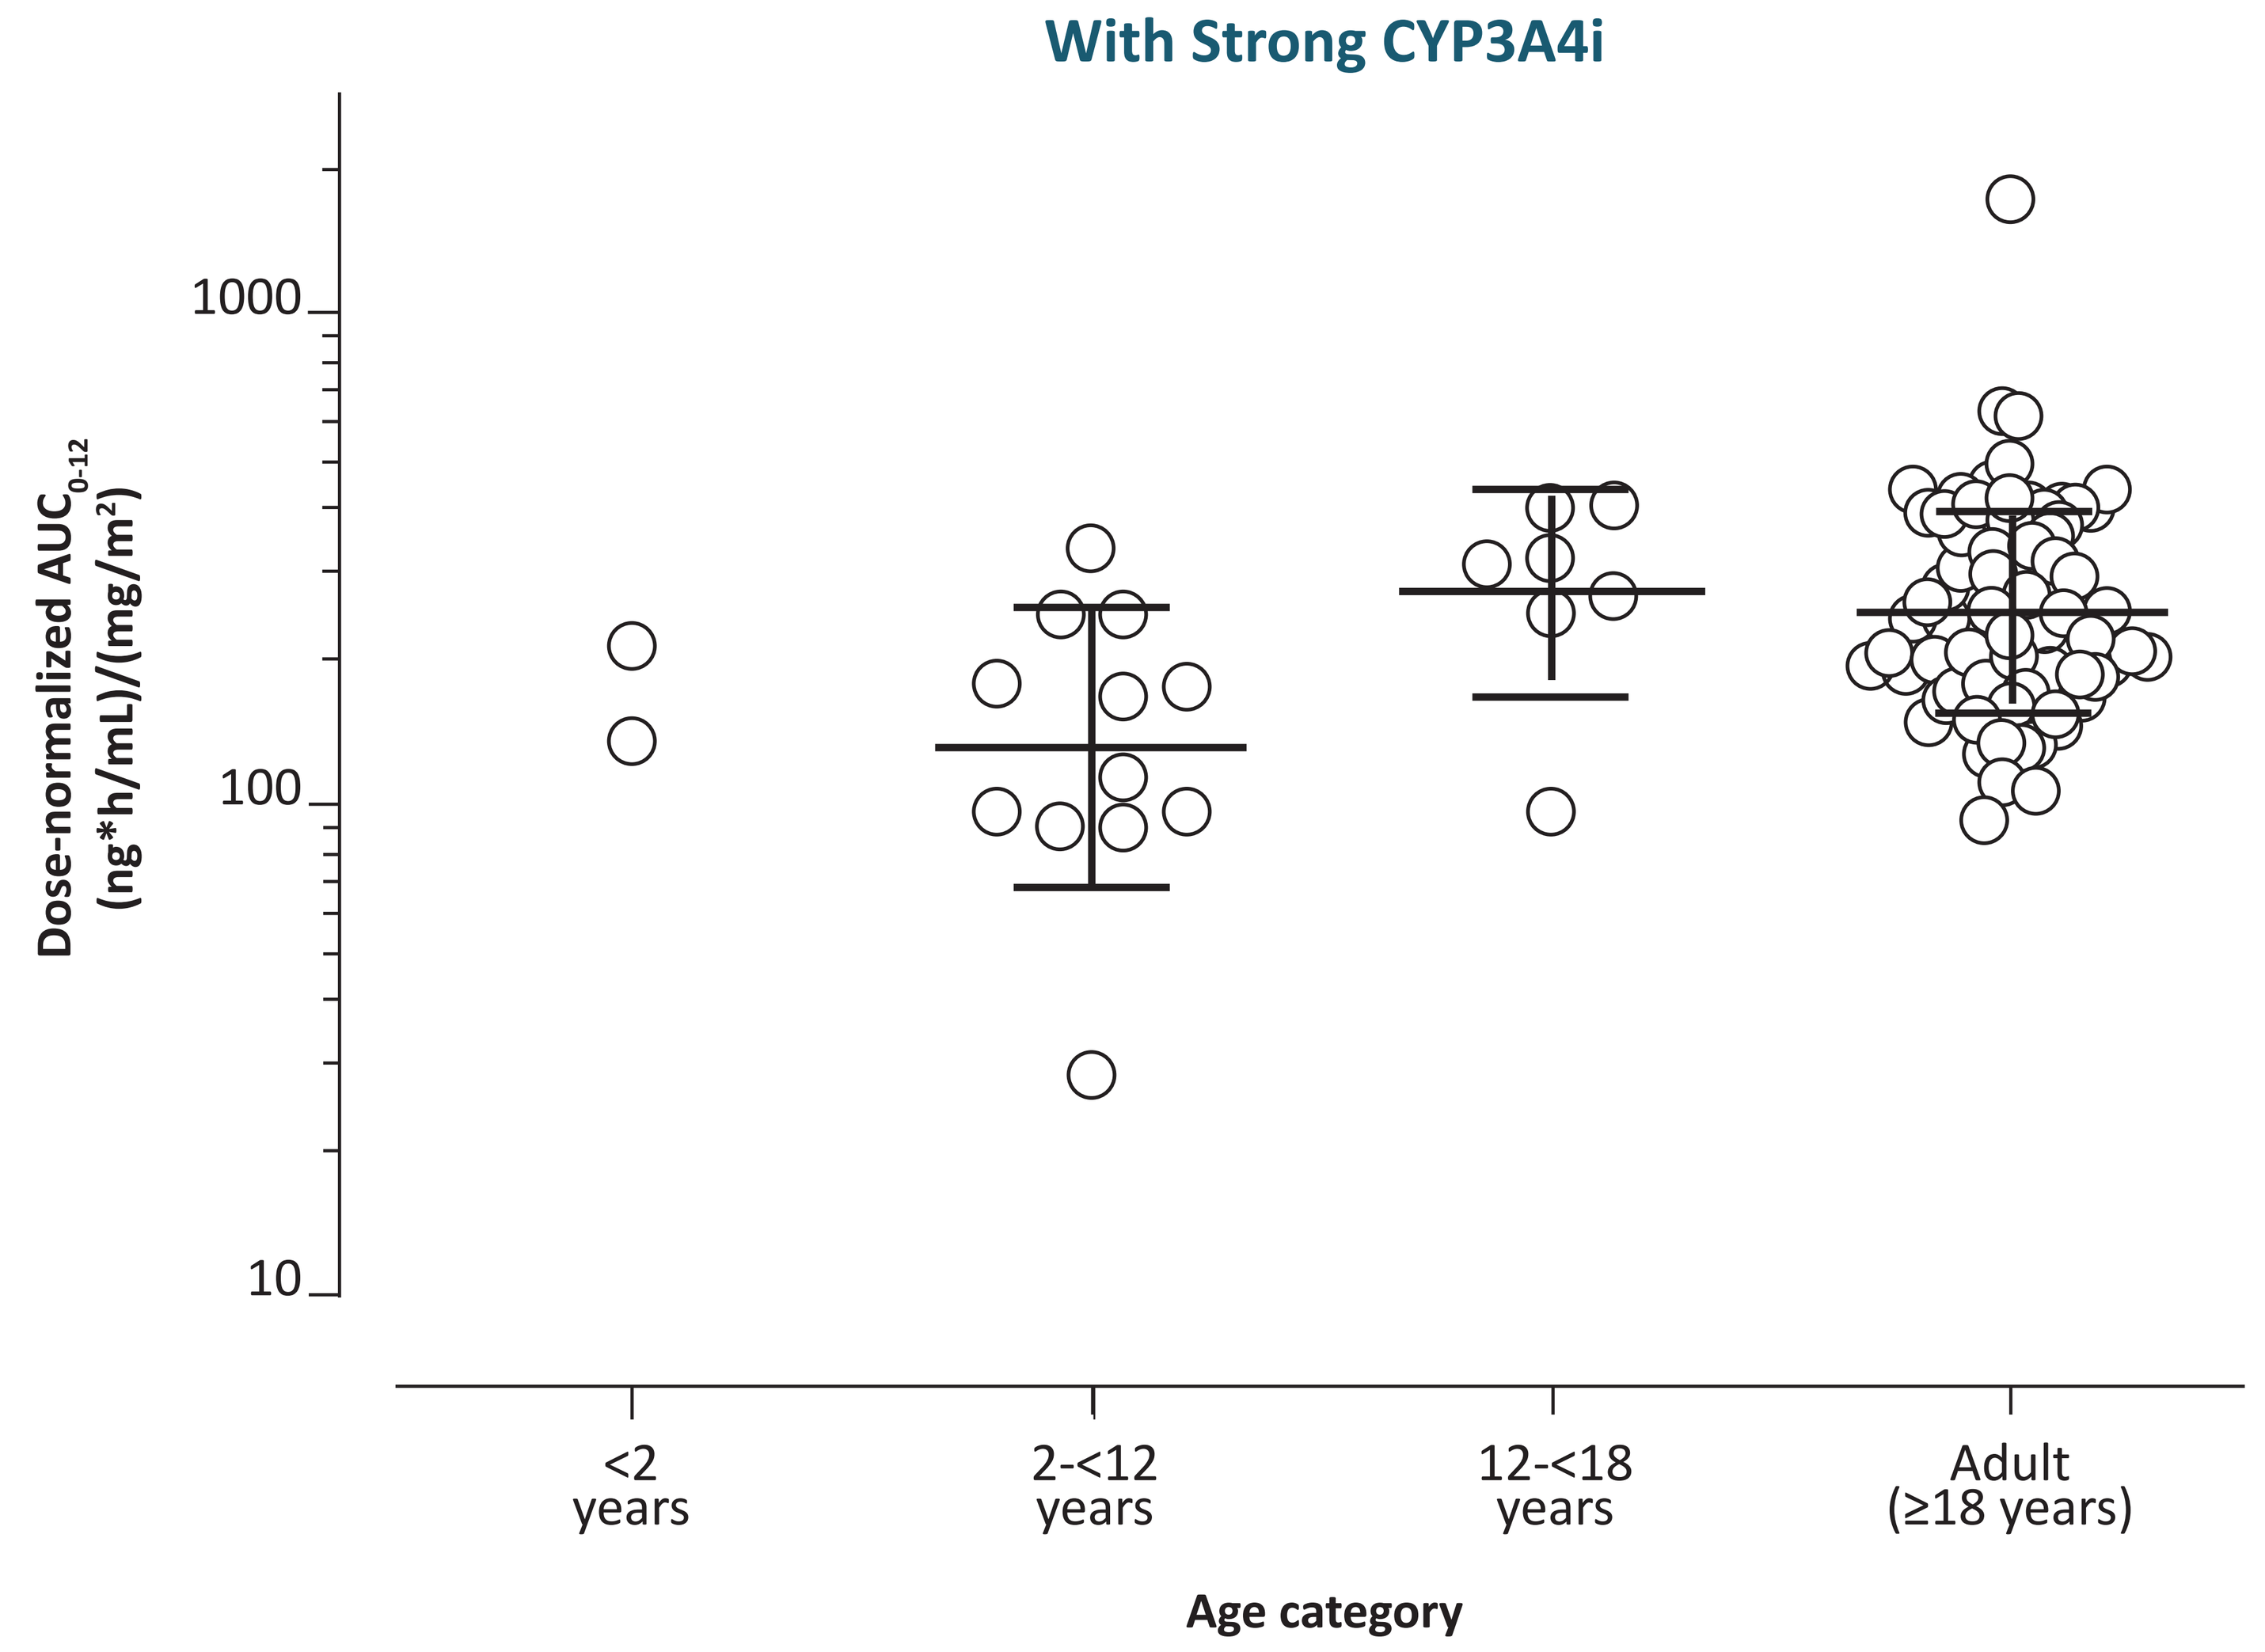
With Strong CYP3A4i
Dose-normalized AUC_{0-12} (ng*h/mL)/(mg/m²)
1000
100
10
<2 years
2-<12 years
12-<18 years
Adult (≥18 years)
Age category

Dose-normalized (BSA-adjusted) AUC_{0-12} of revumenib when given a strong CYP3A4i. Open circles represent individual patients; error bars represent geometric mean and geometric coefficient of variation. AUC, area under the curve; BSA, body surface area; CYP3A4i, cytochrome P450 3A4 inhibitor.

SAFETY

AUGMENT-101

- No dose-limiting toxicities or grade ≥3 treatment-related AEs (TRAEs) were observed
- Grade 2 TRAEs were differentiation syndrome (n=4), decreased appetite, nausea, QTc prolongation, and vomiting (n=1 each)
 - Differentiation syndrome was medically managed with steroids, with no complications reported
- A total of 4 patients discontinued therapy owing to PD and 1 patient withdrew consent

Summary of AEs for Patients in the AUGMENT-101 Trial

AE, n (%)	n=8
Any grade TEAE	8 (100)
Grade ≥3	5 (62.5)
Any grade TRAE	5 (62.5)
Grade ≥3	0
SAE	5 (62.5)
Treatment-related SAE	0
AEs that led to drug discontinuation	0
AEs that led to dose reduction	1 (12.5)
AEs that led to any dose modification[a]	3 (37.5)
AESI[b]	4 (50.0)

AE, adverse event; AESI, adverse event of special interest; CTCAE, common terminology criteria for adverse events; SAE, serious adverse event; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event. [a]Includes decreased, delayed, or skipped dose and drug withdrawn/discontinued. [b]AESI includes differentiation syndrome and prolonged QTc of CTCAE grade 2 or higher.

Revumenib Expanded Access

- Among patients who received expanded access revumenib, available AE data were extracted from the patient narratives as reported by the treating physician
- Investigator-reported TRAEs included thrombocytopenia (n=1), abdominal pain (n=1), differentiation syndrome (n=1), nausea (n=3), neutropenia (n=2), QTc prolongation (n=4), vomiting (n=2), generalized muscle weakness (n=1), neck swelling (n=1), asymptomatic anemia (n=1), bruising (n=1), and intracranial hemorrhage (n=1)
- A total of 15 patients discontinued therapy owing to medical reasons (AE, n=1; no response, n=2; PD, n=10; relapse, n=2), 1 patient died, and 1 patient withdrew consent

CONCLUSIONS

- Among heavily pretreated pediatric patients with acute leukemias who received revumenib monotherapy, encouraging responses were observed among the clinical trial patients (ORR=50%) as well as among the ineligible patients who received revumenib under compassionate use
 - ≥50% of responders in both groups proceeded to transplant, with 1 returning to revumenib as post-HSCT maintenance therapy
- BSA-based dosing strategy of pediatric patients yields exposure similar to that of adults
- Pharmacokinetics of revumenib in combination with strong CYP3A4-inhibiting azole antifungal agents is consistent with that of the adults
- Revumenib demonstrated a manageable safety profile
- Overall, the experience with revumenib highlights the potential clinical benefit of including pediatric patients early in clinical development
- AUGMENT-101 continues to enroll pediatric patients, with pediatric expansion cohorts included in phase 2 with the recommended phase 2 dose of 163 mg (95 mg/m² if <40 kg) every 12 hours with a strong CYP3A4 inhibitor

Acknowledgments: Writing and editorial support were provided under the direction of the authors by Ella A. Kasanga, PhD, PMP®, and Lauren Bragg, ELS, of MedThink SciCom and funded by Syndax Pharmaceuticals, Inc.

References: 1. Wen J, Zhou M, Shen Y, et al. *BMC Cancer.* 2022;22(1):859. **2.** Issa GC, Ravandi F, DiNardo CD, Jabbour E, Kantarjian HM, Andreeff M. *Leukemia.* 2021;35(9):2482-2495. **3.** Quessada J, Cuccuini W, Saultier P, Loosveld M, Harrison CJ, Lafage-Pochitaloff M. *Genes (Basel).* 2021;12(6):924. **4.** Górecki M, Kozioł I, Kopystecka A, Budzyńska J, Zawitkowska J, Lejman M. *Biomedicines.* 2023;11(3):821. **5.** Krivtsov AV, Evans K, Gadrey JY, et al. *Cancer Cell.* 2019;36(6):660-673.e611. **6.** Issa GC, Aldoss I, DiPersio J, et al. *Nature.* 2023;615(7954):920-924.

Syndax

Presented at the 2023 American Society of Pediatric Hematology/Oncology · May 10-13, 2023 · Fort Worth, TX